26588

Patient-reported outcomes from a large, North American-based cohort highlight a greater disease burden for generalized pustular psoriasis versus plaque psoriasis: Real-world evidence from the Corrona Psoriasis Registry

Bruce Strober[1,2], Nirali Kotowsky[3], Rose A. Medeiros[4], Nicole Foster[4], Judson Janak[4], Wendell C. Valdecantos[3], Mary Flack[3], Amanda K. Golembesky[5], Mark Lebwohl[6]

[1]Yale University, New Haven, CT, USA; [2]Central Connecticut Dermatology, Cromwell, CT, USA; [3]Boehringer Ingelheim Pharmaceuticals, Inc., Ridgefield, CT, USA; [4]CorEvitas, LLC, Waltham, MA, USA; [5]Boehringer Ingelheim International GmbH, Ingelheim, Germany; [6]Icahn School of Medicine at Mount Sinai, New York, NY, USA

Background

- Generalized pustular psoriasis (GPP) is a rare, neutrophilic skin disease characterized by episodes of widespread eruption of sterile, macroscopically visible pustules that can occur with or without systemic inflammation[1,2]
- GPP is complex, and often coupled with symptoms such as a high fever and notable comorbidities such as diabetes, respiratory diseases, and hypertension[2,3]
- GPP has a detrimental effect on patient quality of life and can result in mortality; despite this, there are currently no approved treatments in the USA or Canada specifically for GPP[1]
- In addition, due to its rarity, there is limited real-world evidence characterizing GPP[3]
- Establishing the characteristics that distinguish GPP from more common forms of psoriasis (such as plaque psoriasis) may help to inform dermatologists when making treatment decisions, thus improving patient outcomes and facilitating the development of GPP-specific treatments

Methods

- This descriptive, cross-sectional study examined data from the Corrona Psoriasis Registry, a prospective, multicenter, non-interventional registry of patients with psoriasis under the care of a dermatologist in clinical sites throughout the USA and Canada
- Patient sociodemographics, disease characteristics, medication use, and patient-reported outcomes (PROs) at enrollment were examined
- To join the Corrona Psoriasis Registry, patients must have a dermatologist-confirmed diagnosis of psoriasis, be at least 18 years of age, provide written informed consent, and have started or switched to an eligible systemic psoriasis treatment within the past 12 months or at the time of enrollment
- The characteristics of patients with GPP (N=60) were compared with those of patients with plaque psoriasis without pustules (N=4894)
 - These measures were reported as summarized frequencies, percentages, and median and mean visual analog scale (VAS) scores (range 0–100)
- No hypothesis tests were performed
 - The results of this study are purely descriptive

Results

Demographics

- Data from 60 patients with GPP and 4894 patients with plaque psoriasis were included in this study (**Table 1**)

Table 1. Baseline patient demographics and characteristics*

	GPP	Plaque psoriasis
Age, years	N=60	N=4894
median (IQR: 25%, 75%)	48 (40, 63)	52 (40, 61)
mean (SD)	50.9 (14.3)	50.6 (14.3)
Sex	N=60	N=4894
Female	36 (60.0)	2174 (44.4)
Health insurance type†	N=55	N=4662
Private	33 (60.0)	3480 (74.6)
Medicare	16 (29.1)	811 (17.4)
Medicaid	10 (18.2)	546 (11.7)
No insurance	–	153 (3.3)
Education	N=59	N=4882
Bachelor's degree or higher	21 (35.6)	1841 (37.7)
College and/or associate degree	–	1508 (30.9)
High school graduate	23 (39.0)	1189 (24.4)
12th grade or less	–	344 (7.0)
Work status	N=60	N=4886
Full-time	31 (51.7)	2926 (59.9)
Part-time	–	399 (8.2)
Retired	9 (15.0)	781 (16.0)
Disabled	12 (20.0)	369 (7.6)
Other	–	411 (8.4)
Smoking status	N=59	N=4886
Current	12 (20.3)	822 (17.0)
BMI, kg/m²	N=59	N=4826
Normal/underweight (≤24.9)	9 (15.3)	929 (19.2)
Overweight (25.0–29.9)	15 (25.4)	1489 (30.9)
Obese (≥30.0)	35 (59.3)	2408 (49.9)

Not all categories are shown. Categories <5 are masked.
*All values are n (%), unless otherwise stated. †Patients may have more than one insurance type.
BMI, body mass index; IQR, interquartile range; GPP, generalized pustular psoriasis; SD, standard deviation.

- Although there were similarities between patients with GPP and those with plaque psoriasis, some notable differences were observed in the descriptive data from the Corrona Psoriasis Registry

Comorbidities

- A higher proportion of patients with GPP reported anxiety and depression compared with patients with plaque psoriasis (38% vs 26%, as identified in the EQ-5D-3)
 - More patients with GPP than plaque psoriasis had comorbid conditions such as asthma (12% vs 6%), infections (48% vs 27%), and diabetes (22% vs 14%) (**Figure 1**)

Figure 1. Occurrence of specific comorbidities in patients with GPP and plaque psoriasis

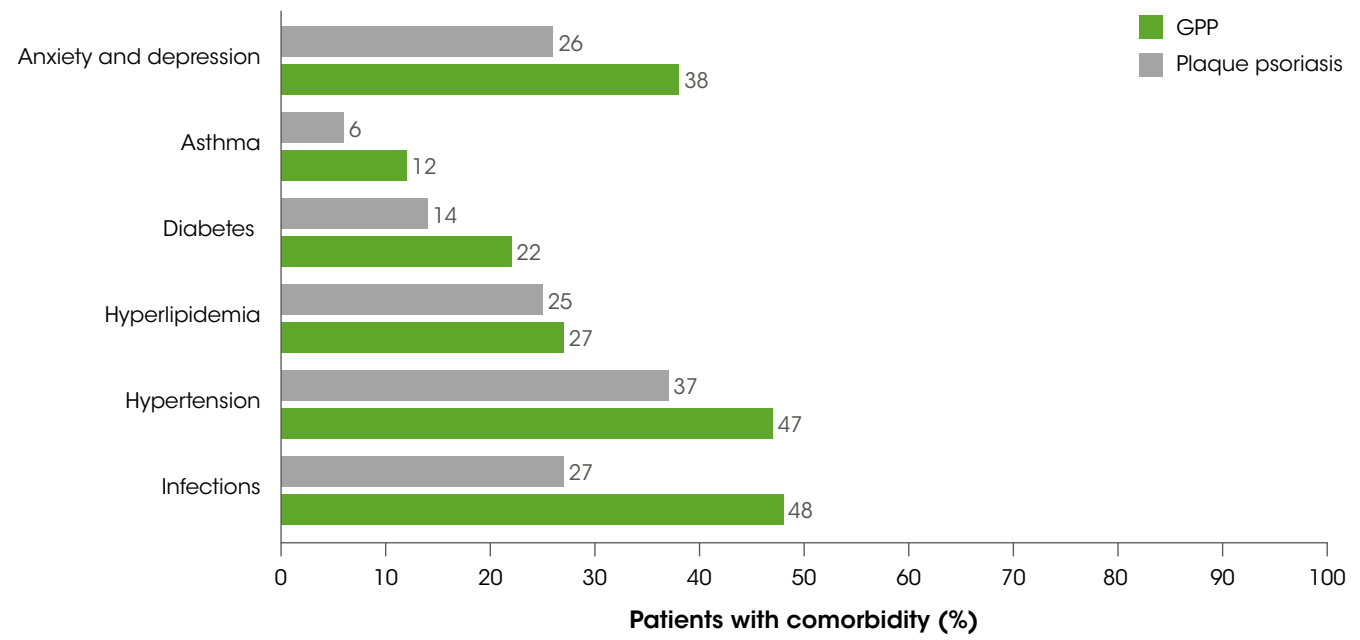

Anxiety and depression
26
38
Asthma
6
12
Diabetes
14
22
Hyperlipidemia
25
27
Hypertension
37
47
Infections
27
48
0 10 20 30 40 50 60 70 80 90 100
Patients with comorbidity (%)
GPP
Plaque psoriasis

GPP, generalized pustular psoriasis.

PROs and quality of life

- Patients with GPP reported higher median (25th percentile, 75th percentile) and mean (standard deviation [SD]) VAS scores than those with plaque psoriasis (respectively) for:
 - Itch: median 59 (10, 85) vs 22 (5, 70); mean 47.7 (36.8) vs 35.4 (34.3)
 - Fatigue: median 44 (15, 73) vs 20 (4, 50); mean 42.6 (31.2) vs 29.5 (28.4)
 - Pain: median 20 (3, 62) vs 5 (0, 35); mean 33.1 (34.2) vs 21.5 (29.0)
- Median (25th percentile, 75th percentile) and mean (SD) VAS scores for patient global assessment (a PRO measure of global disease impact from 0 to 100, for which a higher number indicates greater impact) were higher in patients with GPP compared to patients with plaque psoriasis (median 50 [15, 74] vs 30 [10, 60]; mean 45.6 [31.2] vs 35.9 [30.1])
- Median (25th percentile, 75th percentile) values are displayed in **Figure 2**

Figure 2. Patient-reported symptom measures (median VAS score, range 1–100) for GPP and plaque psoriasis

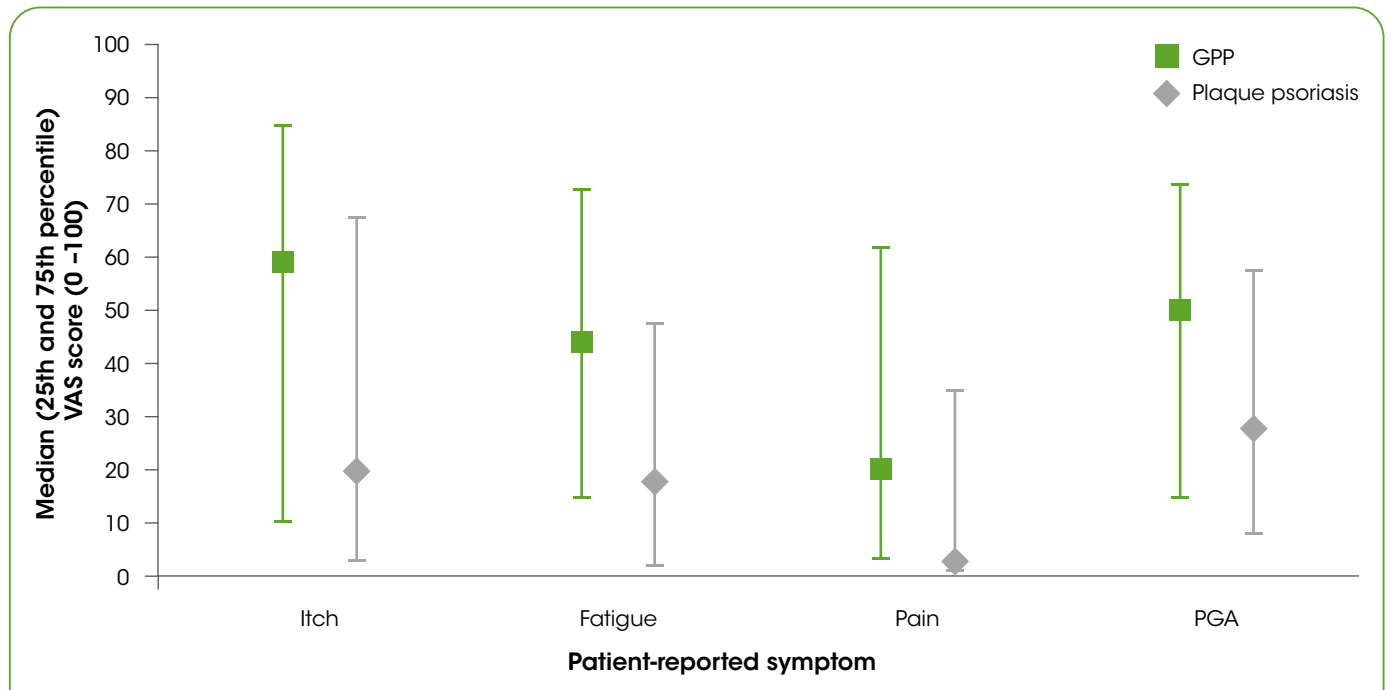

Median (25th and 75th percentile) VAS score (0 –100)
100
90
80
70
60
50
40
30
20
10
0
Itch
Fatigue
Pain
PGA
Patient-reported symptom
GPP
Plaque psoriasis

Whiskers on graph represent the 25th and 75th percentiles. A higher VAS score indicates worse symptoms.
GPP, generalized pustular psoriasis; PGA, patient global assessment; VAS, visual analog scale.

- Median (25th percentile, 75th percentile) and mean (SD) percentage of patients who reported impairment while working were higher in patients with GPP compared with those with plaque psoriasis (median 24 [3, 50] vs 0 [0, 15]; mean 28.6 [26.2] vs 12.5 [21.3])
 - Similarly, median (25th percentile, 75th percentile) and mean (SD) percentage of patients who reported impairment in daily activities were higher in patients with GPP compared with patients with plaque psoriasis (median 20 [0.5, 55] vs 3 [0, 25]; mean 31.9 (32.9) vs 17.1 (25.5)

Treatment experience

- Patients with GPP had more treatment experience: 15% had received ≥2 systemic agents versus 7% of patients with plaque psoriasis

Discussion

Limitations

- There are some evident limitations to this study
 - The specific registry inclusion criteria and descriptive nature of this study mean that the results apply only to this registry sample rather than the general population
 - Furthermore, the patients included in the Corrona Psoriasis Registry may not represent typical patients with recurring GPP flares
 - Patients were classified into the GPP cohort based on their full history of psoriasis subtypes
 - Thus, it is unclear whether a 'patient with GPP' had pustules at the time of enrollment (when outcome measures were collected); as a result, symptom severity for patients with GPP may have been underestimated
 - Furthermore, the Corrona Psoriasis Registry only contains information on patients in North America and therefore may not be representative of GPP on a global scale

Conclusions

- The results of this descriptive study comparing patients with GPP with patients with plaque psoriasis without pustules suggest that GPP and plaque psoriasis without pustules have differing symptom severity and impact on quality of life
- In this sample, patient-reported VAS measures of itching, pain, and fatigue were more severe in patients with GPP than those with plaque psoriasis
 - This may explain the data showing that a higher proportion of patients with GPP than those with plaque psoriasis experienced impairment while working and performing daily activities
- In this sample, patients with GPP also reported more anxiety and depression than patients with plaque psoriasis
- Descriptively, notable comorbidities such as asthma, infections, and diabetes were more common in patients with GPP than those with plaque psoriasis
- These observed differences in the characteristics and patient-reported outcomes of patients with GPP could help to guide the development of future treatments and further delineate the diagnostic criteria
- Furthermore, patients with GPP in this sample were prescribed more therapies than patients with plaque psoriasis, which may suggest that current treatment options for GPP do not adequately resolve the disease, highlighting the need to develop GPP-specific treatments

References

1. Benjegerdes KE, et al. *Psoriasis (Auckl)* 2016;6:131–144.
2. Uppala R, et al. *Cell Mol Immunol* 2020;18:307–317.
3. Ly K, et al. *Psoriasis (Auckl)* 2019;9:37–42.

Disclosures

This study design and conduct were the result of a collaborative effort between CorEvitas, LLC and Boehringer Ingelheim, and financial support was provided by Boehringer Ingelheim International GmbH, Ingelheim, Germany. B Strober reports that he is a consultant (honoraria) for AbbVie, Almirall, Amgen, Arcutis, Arena, Aristea, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Dermavant, Dermira, Equillium, Janssen, Leo Pharmaceuticals, Eli Lilly, Meiji Seika Pharma, Mindera, Novartis, Pfizer, GlaxoSmithKline, UCB Pharma, Sun Pharmaceuticals Industries, Ortho Dermatologics, Regeneron, and Sanofi-Genzyme; a speaker for AbbVie, Amgen, Eli Lilly, Janssen, and Ortho Dermatologics; a scientific director (consulting fee) for the Corrona Psoriasis Registry; and an investigator for Dermavant, AbbVie, Corrona Psoriasis Registry, Dermira, Cara Therapeutics, and Novartis. M Lebwohl reports that he is an employee of Mount Sinai and receives research funds from AbbVie, Amgen, Arcutis, Boehringer Ingelheim, Dermavant, Eli Lilly, Incyte, Janssen Research & Development, Leo Pharmaceuticals, Ortho Dermatologics, Pfizer, and UCB, and a consultant for Aditum Bio, Allergan, Almirall, Arcutis, Avotres Therapeutics, BirchBioMed, BMD Skincare, Boehringer Ingelheim, Bristol Myers Squibb, Cara Therapeutics, Castle Biosciences, CorEvitas, Dermavant Sciences, Evelo, Facilitate International Dermatologic Education, Foundation for Research and Education in Dermatology, Inozyme Pharma, Kyowa Kirin, Leo Pharmaceuticals, Meiji Seika Pharma, Menlo, Mitsubishi, Neuroderm, Pfizer, Promius/Dr. Reddy's Laboratories, Serono, Theravance, and Verrica. N Kotowsky, WC Valdecantos, and M Flack are full-time employees of Boehringer Ingelheim. AK Golembesky reports being an employee of Boehringer Ingelheim at the time of the study being conducted, but is currently an employee of GlaxoSmithKline. RA Medeiros, N Foster, and J Janak are employees of CorEvitas, LLC. The Corrona Psoriasis Registry is sponsored by CorEvitas, LLC. CorEvitas, LLC has been supported through contracted subscriptions in the last 2 years by AbbVie, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Eli Lilly and Company, Genentech, Gilead, Janssen, Momenta, Novartis, Ortho Dermatologics, Pfizer, Regeneron, Sanofi, and Sun Pharmaceutical Industries.

Acknowledgments

Editorial assistance in the preparation of this poster was provided by Maria Thomas and Imogen Allred, PhD, of OPEN Health Communications (London, UK) and funded by Boehringer Ingelheim.

Click the icon to access an interactive microsite for this Smart poster

Scan QR code for an interactive, electronic device-friendly copy of the poster https://bit.ly/3qnCfSC

American Academy of Dermatology Virtual Meeting Experience, April 23–25, 2021